>66,000 Pregnancies Screened Between 8-10 Weeks Gestation: The Impact of Fetal Fraction Amplification on Prenatal Cell-Free DNA Screening

Jhett Bordwell, PhD;[1] Summer Pierson, MS, CGC;[1] Katie Hunt, MS, CGC;[1] Cathy Wicklund, MS, CGC;[1] Katie Johansen Taber, PhD;[1] Dale Muzzey, PhD[1]

1 Myriad Genetics, Inc., Salt Lake City, UT

Myriad genetics®

Purpose

- Performance of prenatal cell-free DNA (pcfDNA) screening at gestational ages <10 weeks has been challenging to implement due in part to the lower fetal fractions (FF) at earlier gestational ages impacting screen failure rates.
- Fetal fraction amplification (FFA) enables reliable screening at gestational ages as early as 8 weeks while maintaining low failure rates[1].
- Here we describe the lab experience for over 66,000 screens performed for pregnancies in the early gestational age period (EGA) of 8w0d to 9w6d.

Methods

- Data from pcfDNA screens completed between 11/19/2024 and 03/31/2026 were considered for analysis. The following were reported:
 - Number of orders received
 - Screen-positive results
 - Screen failures
- FF medians and first and third quartiles (Q1, Q3) for the 8- and 9-week gestational age period were calculated.
- Where provided, BMI information was analyzed.

1. Dugoff L, Mabey B, Bordwell J, Pierson S, Johansen Taber K, Muzzey D. Prenatal Cell-Free DNA Screening With Fetal Fraction Amplification at 6-9 Weeks of Gestation. Obstet Gynecol. 2026 Aug 1;148(2):149-156. doi: 10.1097/AOG.0000000000006324. Epub 2026 May 21. PMID: 42166774; PMCID: PMC13361953.

Results

- 66,643 orders were completed for patients between 8w0d and 9w6d.
 - 1,645 (2.47%) were screen-positive (**Table 1**).
 - Of 647 rare autosomal aneuploidies, 563 were trisomies.
 - 50 samples were screen-positive for two or more autosomal abnormalities.
 - Results could not be generated for 238 samples, resulting in a screen-failure rate of 0.36% (**Table 2 and Table 3**); 64 samples (0.1%) failed due to insufficient FF (**Table 2**).

Table 1. Screen positive results of week 8 and 9 combined

Result type	1,645 screen-positive reports (2.47% screen-positive rate)
Aneuploidies	
Trisomy 21	**341** (20.73%)
Trisomy 18	**121** (7.36%)
Trisomy 13	**103** (6.26%)
Sex chromosome aneuploidy (SCA)	**531** (32.38%)
Rare autosomal aneuploidies (RAA)	**647** (39.33%)
Microdeletions	
1p36	**14** (0.85%)
22q11.2	**9** (0.55%)
15q11.2	**8** (0.49%)
5p	**7** (0.43%)
4p	**3** (0.18%)

- Median FF across the cohort was 15.0% (Q1=11.0%; Q3=19.8%). For samples in the 8th week of pregnancy, median FF was 13.4% (Q1=9.9%; Q3=17.7%) and for samples in the 9th week of pregnancy, median FF was 16.8% (Q1=12.5%; Q3=21.5%) (**Figure 1**).

Table 2. Screen failure rates for weeks 8 and 9 combined

Reason for failure	238 total no call reports
Insufficient FF	**64 Samples** (0.10%)
Other QC metrics	**174 Samples** (0.26%)

0.36% overall screen failure rate

Figure 1. Fetal Fraction distribution at 8 and 9 weeks

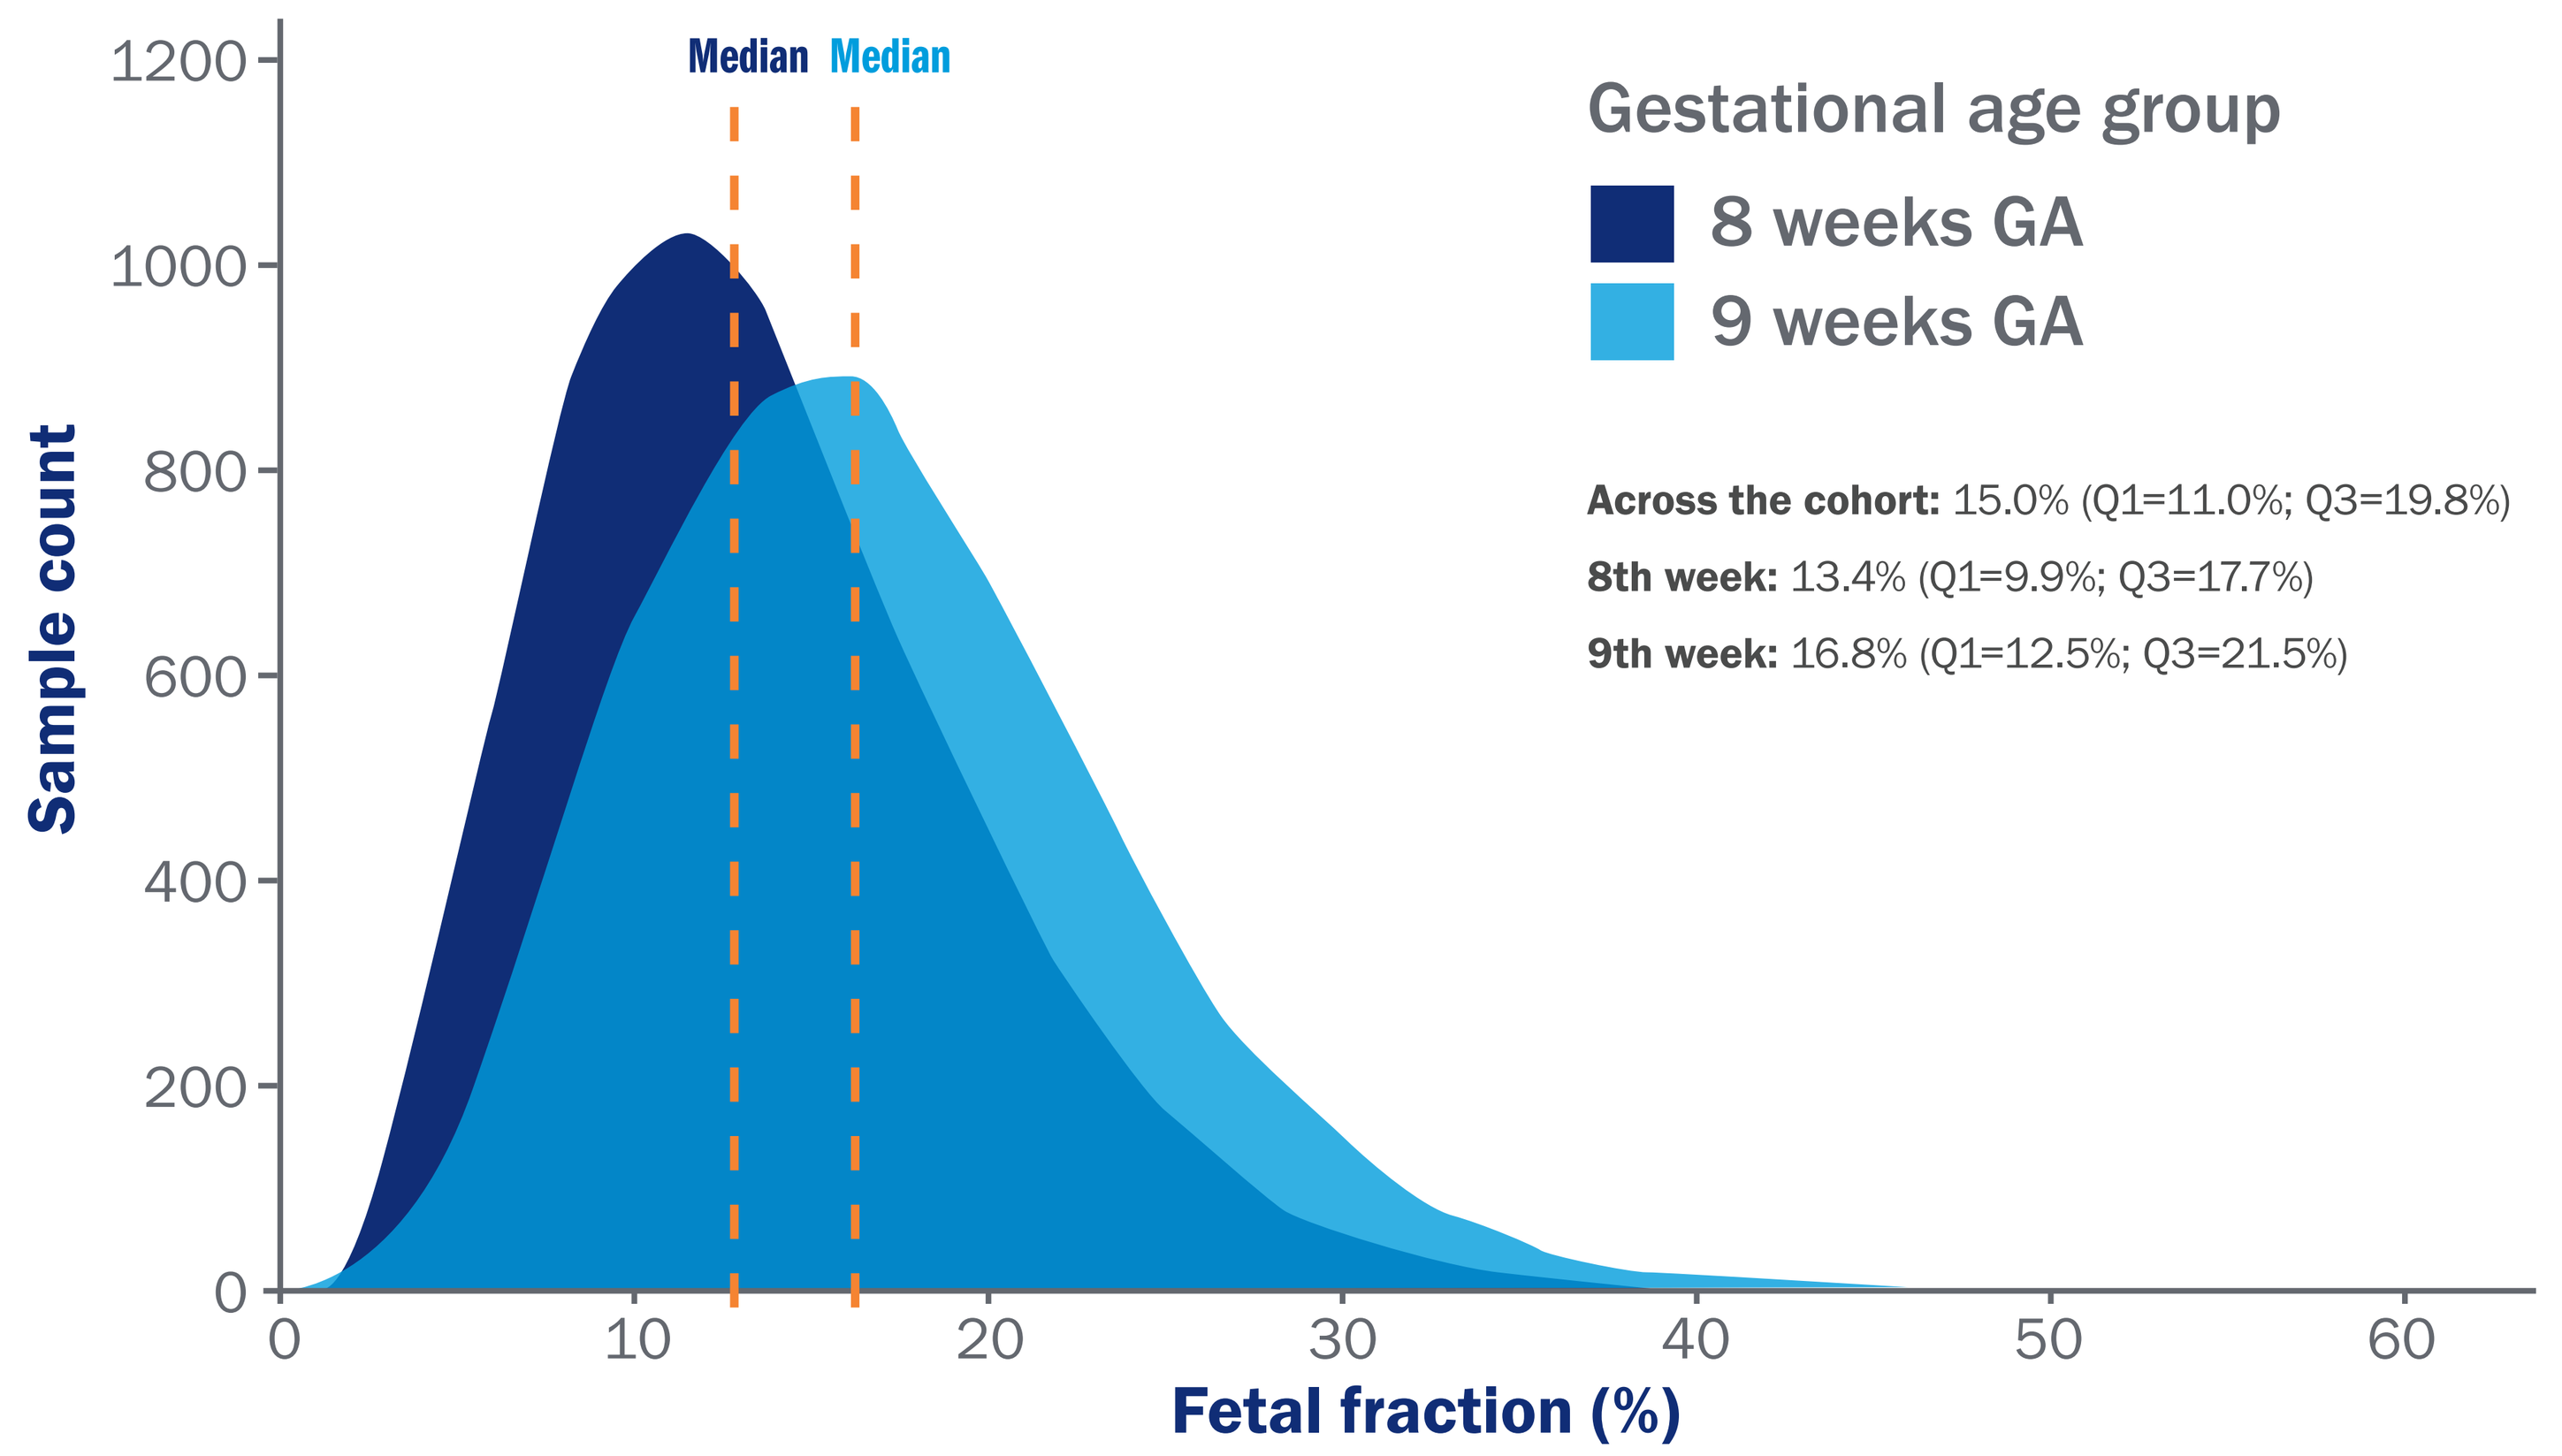

Median
Median
Gestational age group
8 weeks GA
9 weeks GA
Across the cohort: 15.0% (Q1=11.0%; Q3=19.8%)
8th week: 13.4% (Q1=9.9%; Q3=17.7%)
9th week: 16.8% (Q1=12.5%; Q3=21.5%)
Sample count
Fetal fraction (%)

- For samples in which BMI could be assessed (n=40,443), the proportion at BMI ≥30 was 34.5% (n=13,938). For those with BMI ≥30, median FF was 12.1% (Q1=8.9%; Q3=15.9%) (**Figure 2**).

Table 3. Failures due to insufficient FF

Week	n (Rate)
Week 8	**48 Samples** (0.07%)
Week 9	**16 Samples** (0.02%)

Figure 2. Fetal Fraction distribution at high BMI ≥30

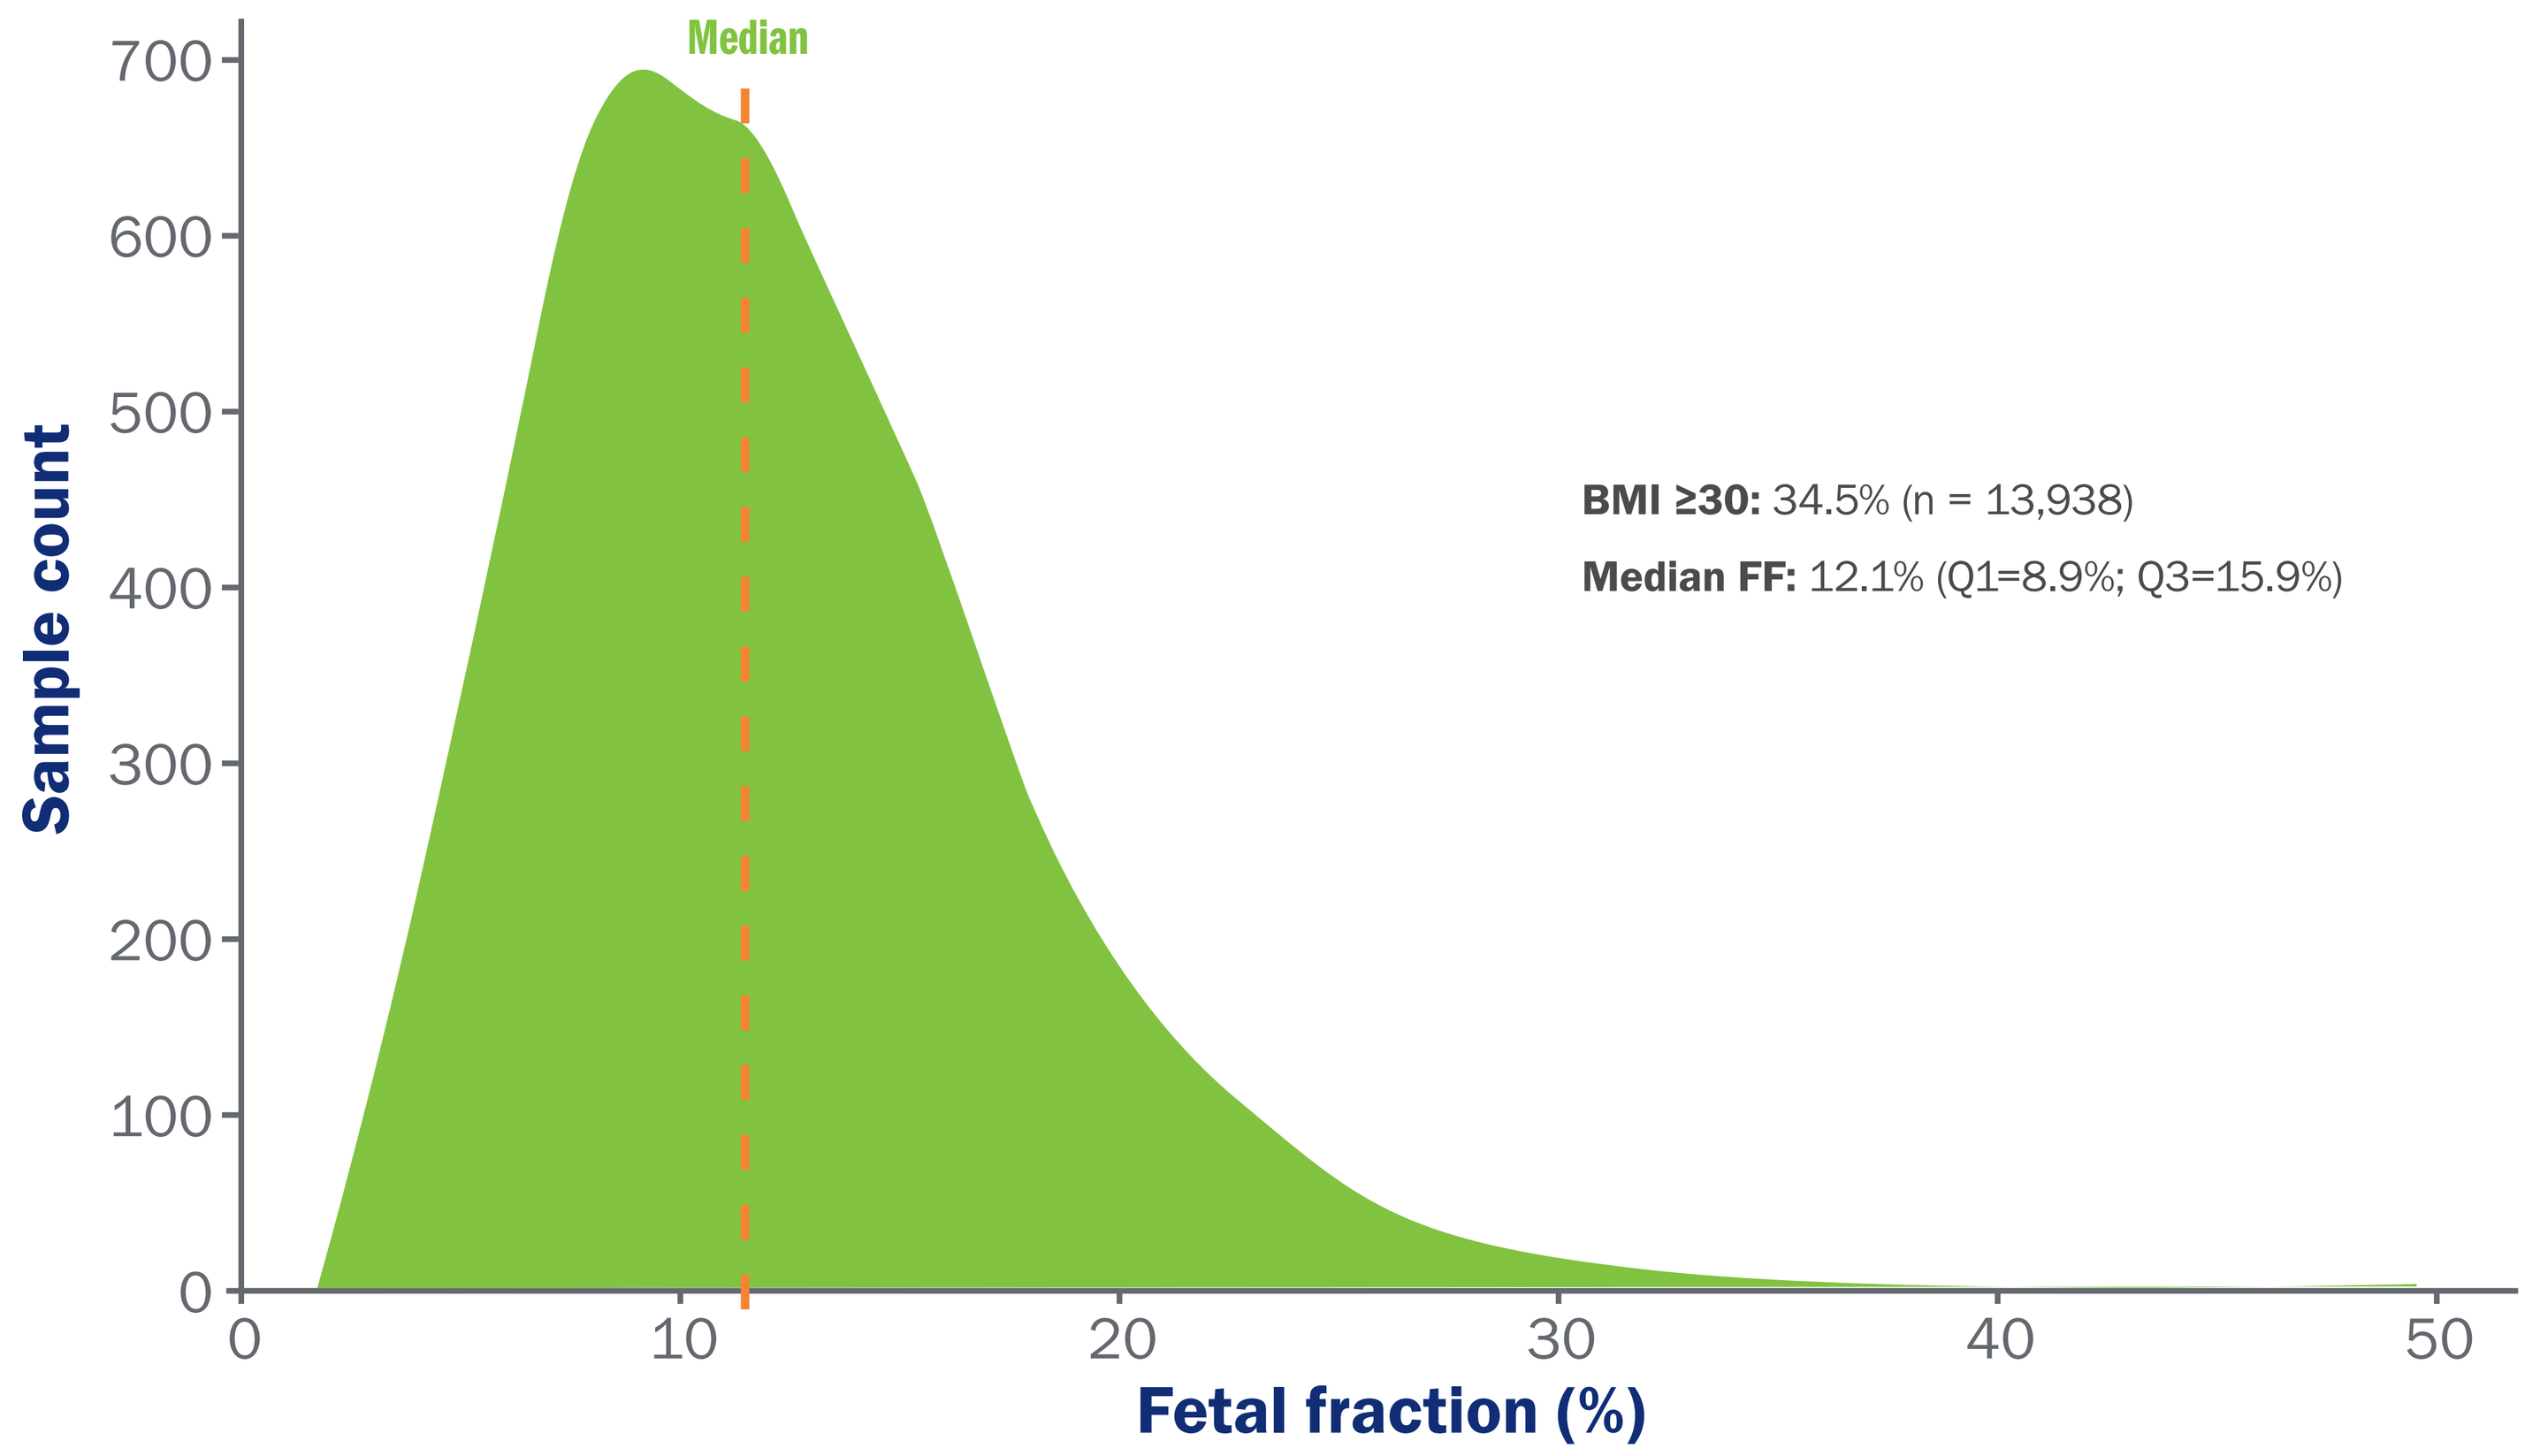

Median
BMI ≥30: 34.5% (n = 13,938)
Median FF: 12.1% (Q1=8.9%; Q3=15.9%)
Sample count
Fetal fraction (%)

Conclusions

- This pcfDNA assay with FFA demonstrates sufficient FF for pregnancies within the EGA period as well as a low FF failure of 0.1%, including pregnancies with higher BMIs.
- pcfDNA screening at an EGA offers a viable option for individuals who desire information earlier in pregnancy than what is typically available.

Disclosures: All authors were employees of Myriad Genetics, Inc. at the time of this study and received salaries and stock as compensation.

Email for questions: **cathy.wicklund@myriad.com**

I-2026-6