Prevalence of Elevated Breast Cancer Risk in Unaffected Menopausal-age Women to Potentially Inform Hormone Therapy Initiation

Edith Smith, DNP, CNM, WHNP-BC; Sarah Ratzel, PhD; Claire Miller, MS

Myriad Genetics, Salt Lake City, Utah

Myriad genetics®

Background

- Hormone therapy (HT) has been shown to relieve vasomotor symptoms (VMS) of menopause and is FDA-approved as first line therapy for the treatment of moderate to severe symptoms.
- The ideal candidates for HT are women under age 60 (or within 10 years of menopause onset) with no contraindications to HT use.
- HT use can increase breast cancer (BC) risk, however, and those at increased risk may be better suited for non-hormonal therapies for treatment of menopause-related VMS.
- Guidelines recommend comprehensive BC risk assessment be performed prior to initiation of HT and discourage HT use in high-risk individuals.
- The aim of this study was to characterize the classification of BC risk, as calculated by Tyrer-Cuzick (TC) and/or a combined risk score (CRS), in unaffected menopausal-age women (ages 50-60 years old) who underwent multigene panel testing and were found not to harbor a pathogenic variant (PV) in a BC-associated gene.

Methods

- Evaluated a sequential cohort of individuals undergoing multigene panel testing for hereditary cancer risk between January 2021 and January 2026
- Myriad Genetics performed germline genetic testing and provided BC risk estimates using TC and a polygenic-based risk estimate that combines a polygenic risk score with TC (CRS)
 - High risk defined as ≥20% estimated lifetime risk of BC
- Descriptive statistics were used to characterize the number of individuals with a high-risk result

Figure 1. Multigene Panel and CRS

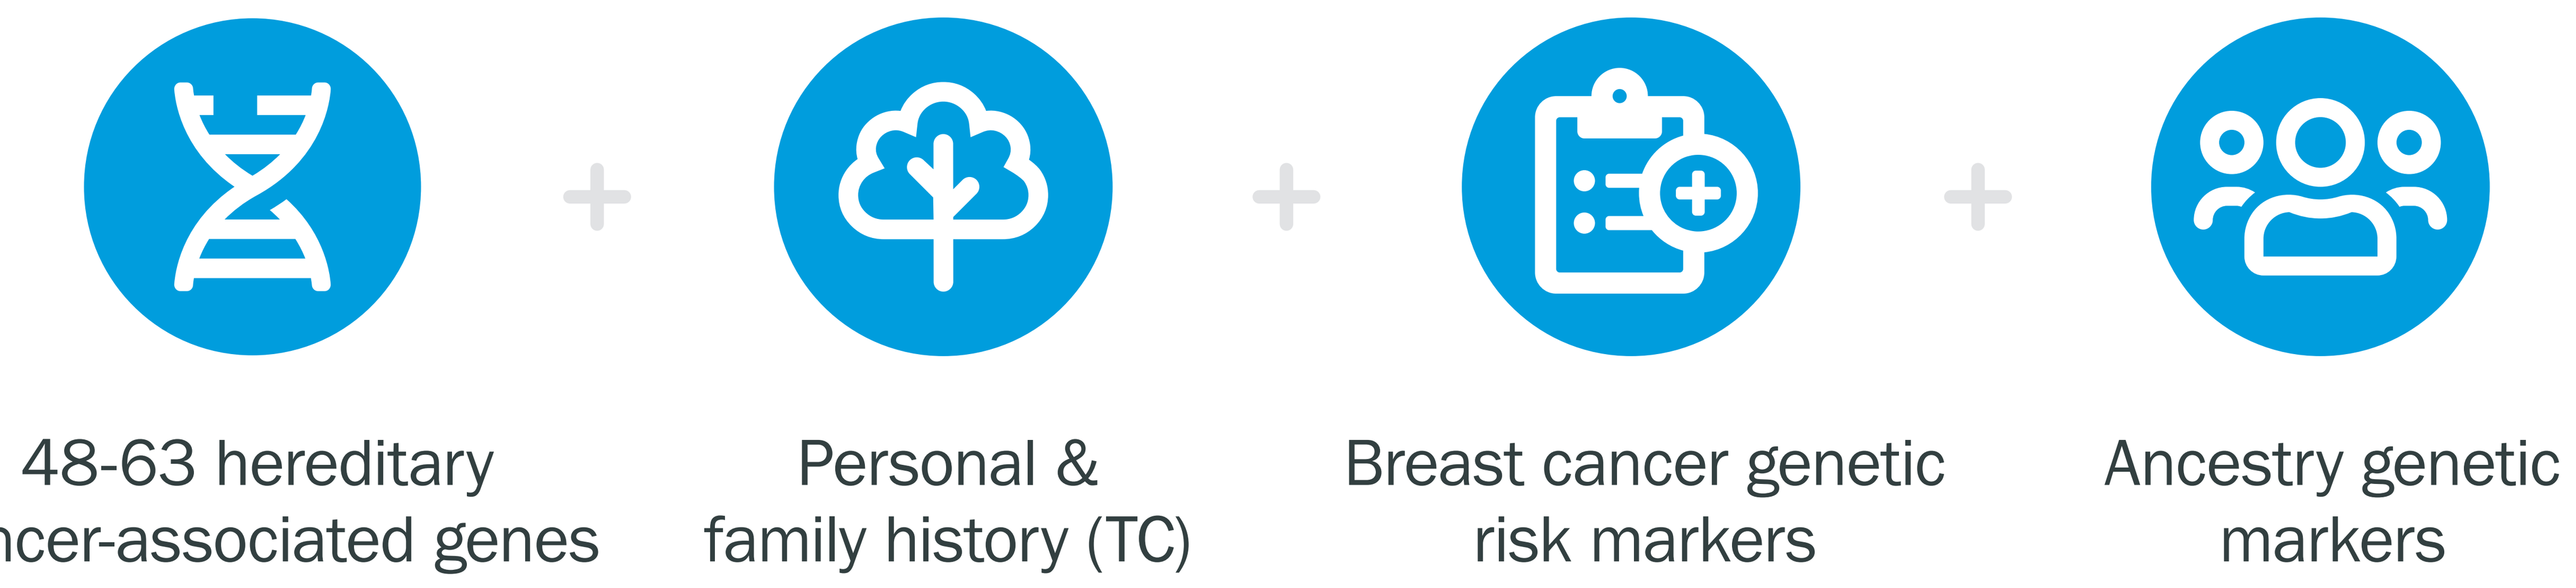
48-63 hereditary cancer-associated genes
+
Personal & family history (TC)
+
Breast cancer genetic risk markers
+
Ancestry genetic markers

Results

Figure 2. Percentage of Patients Identified with/without a BC-associated PV (N=150,604)

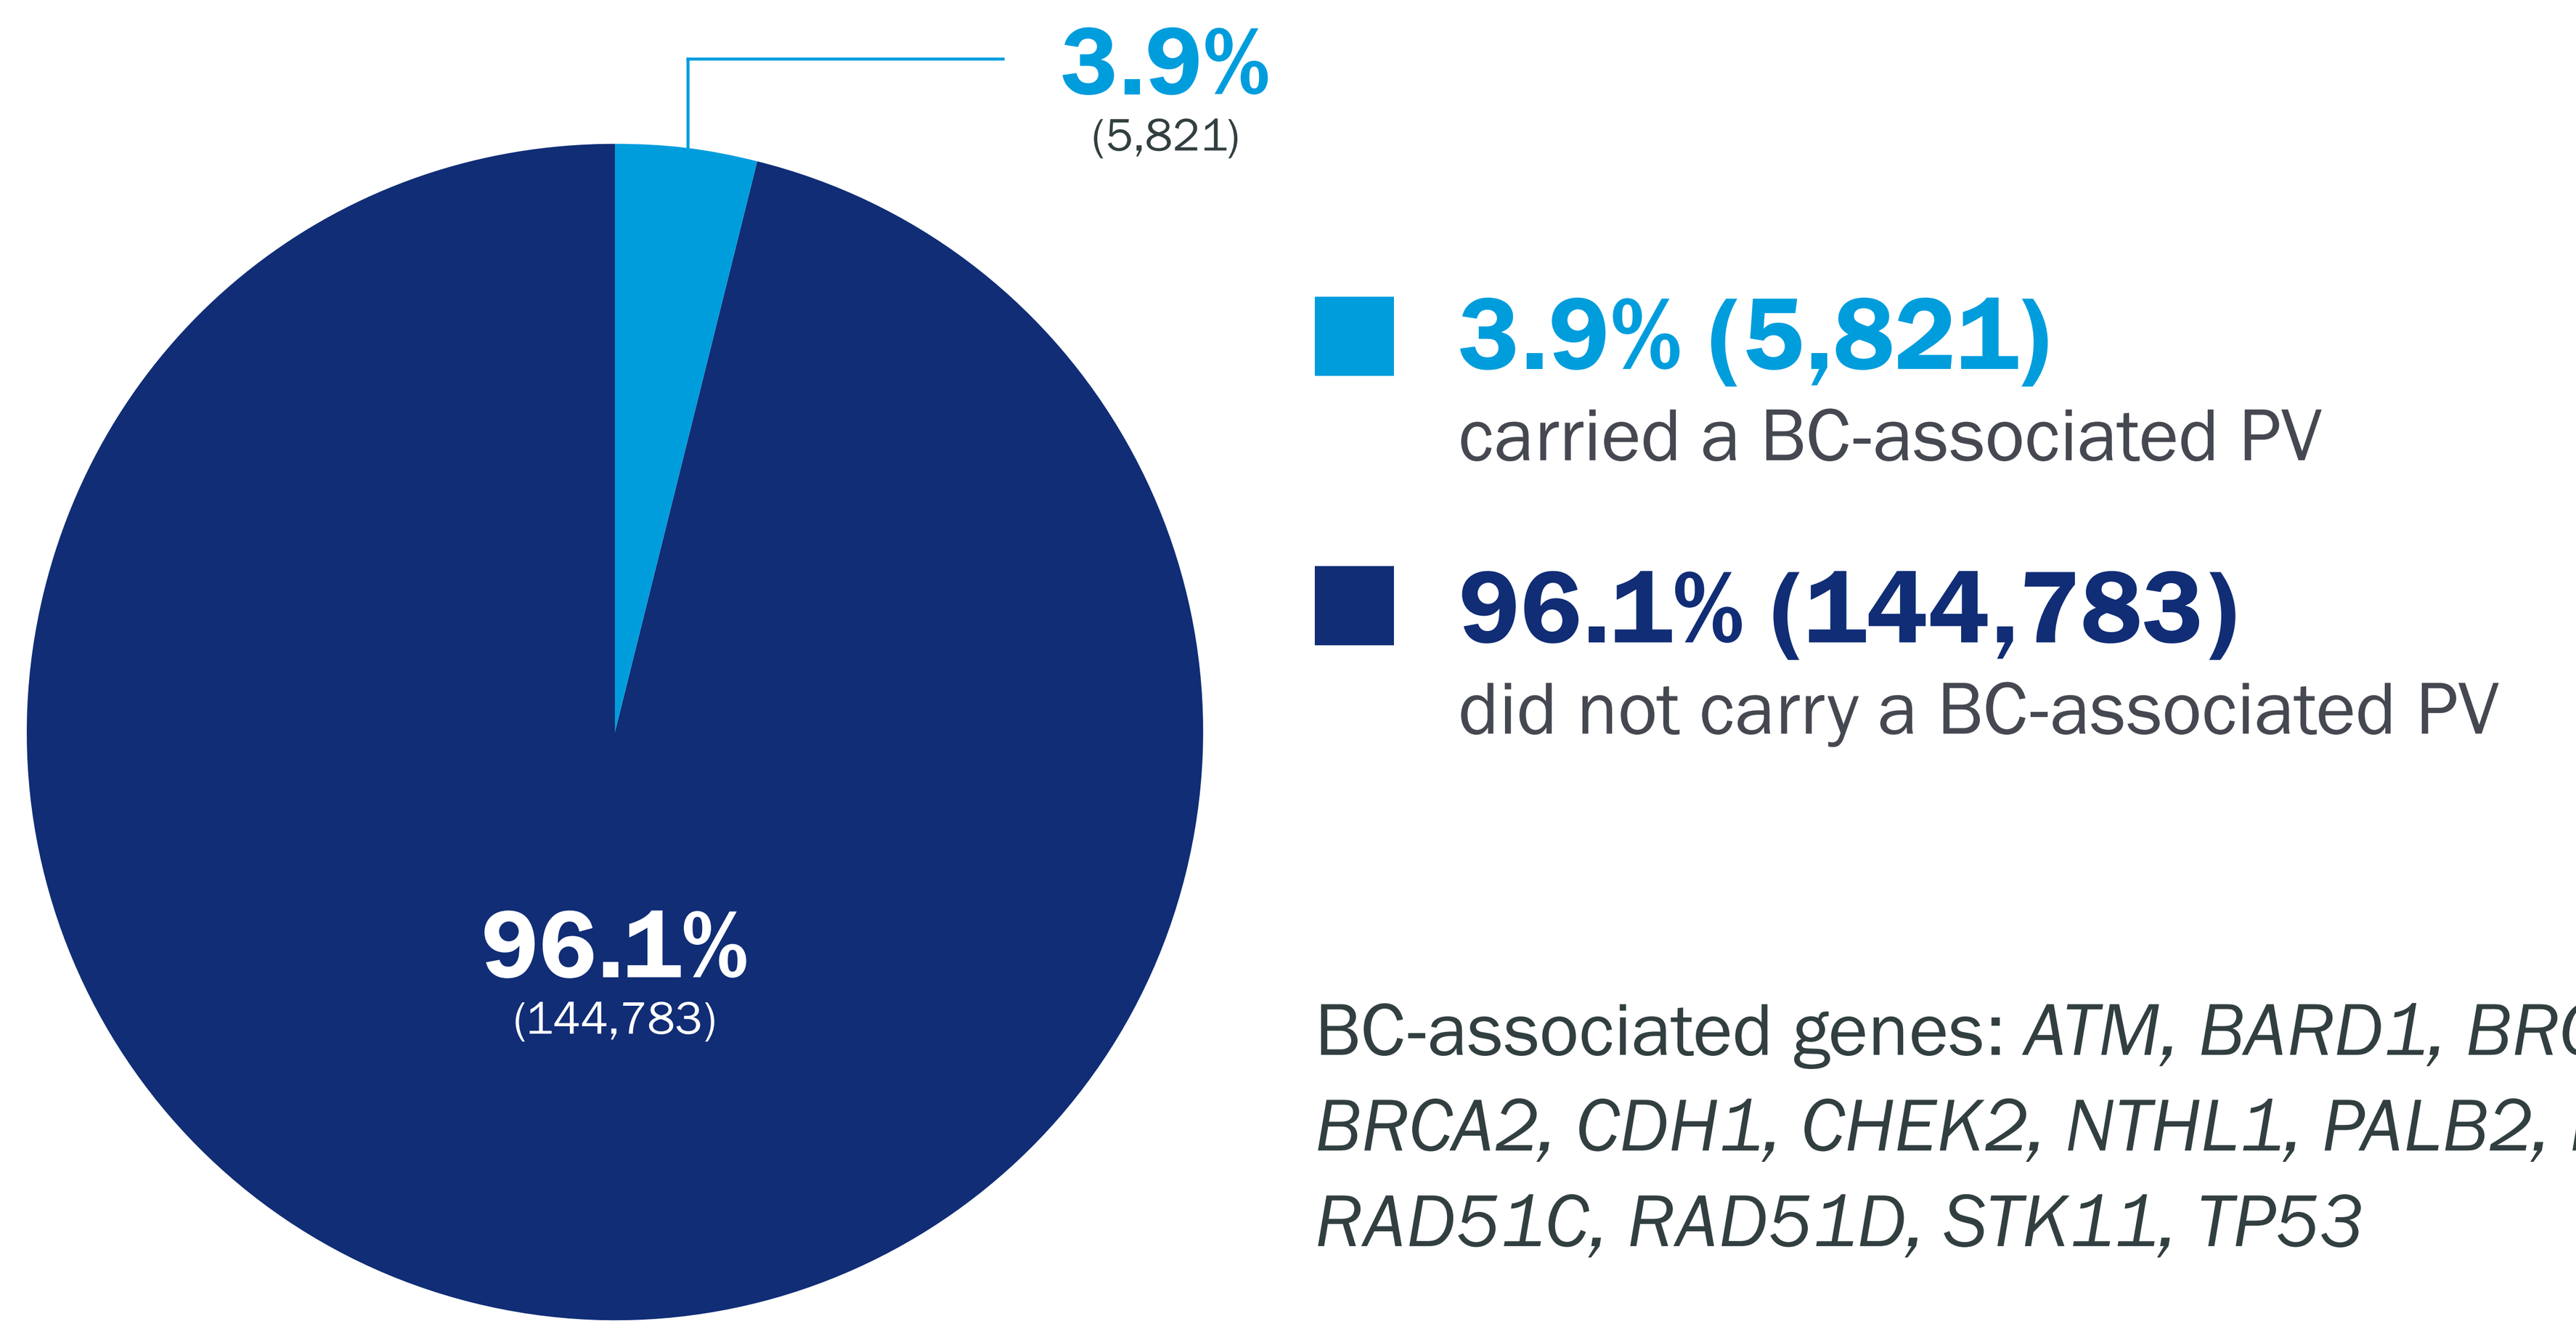
3.9%
(5,821)
96.1%
(144,783)
3.9% (5,821)
carried a BC-associated PV
96.1% (144,783)
did not carry a BC-associated PV

BC-associated genes: *ATM, BARD1, BRCA1, BRCA2, CDH1, CHEK2, NTHL1, PALB2, PTEN, RAD51C, RAD51D, STK11, TP53*

Figure 3. Breast Cancer Risk Classification

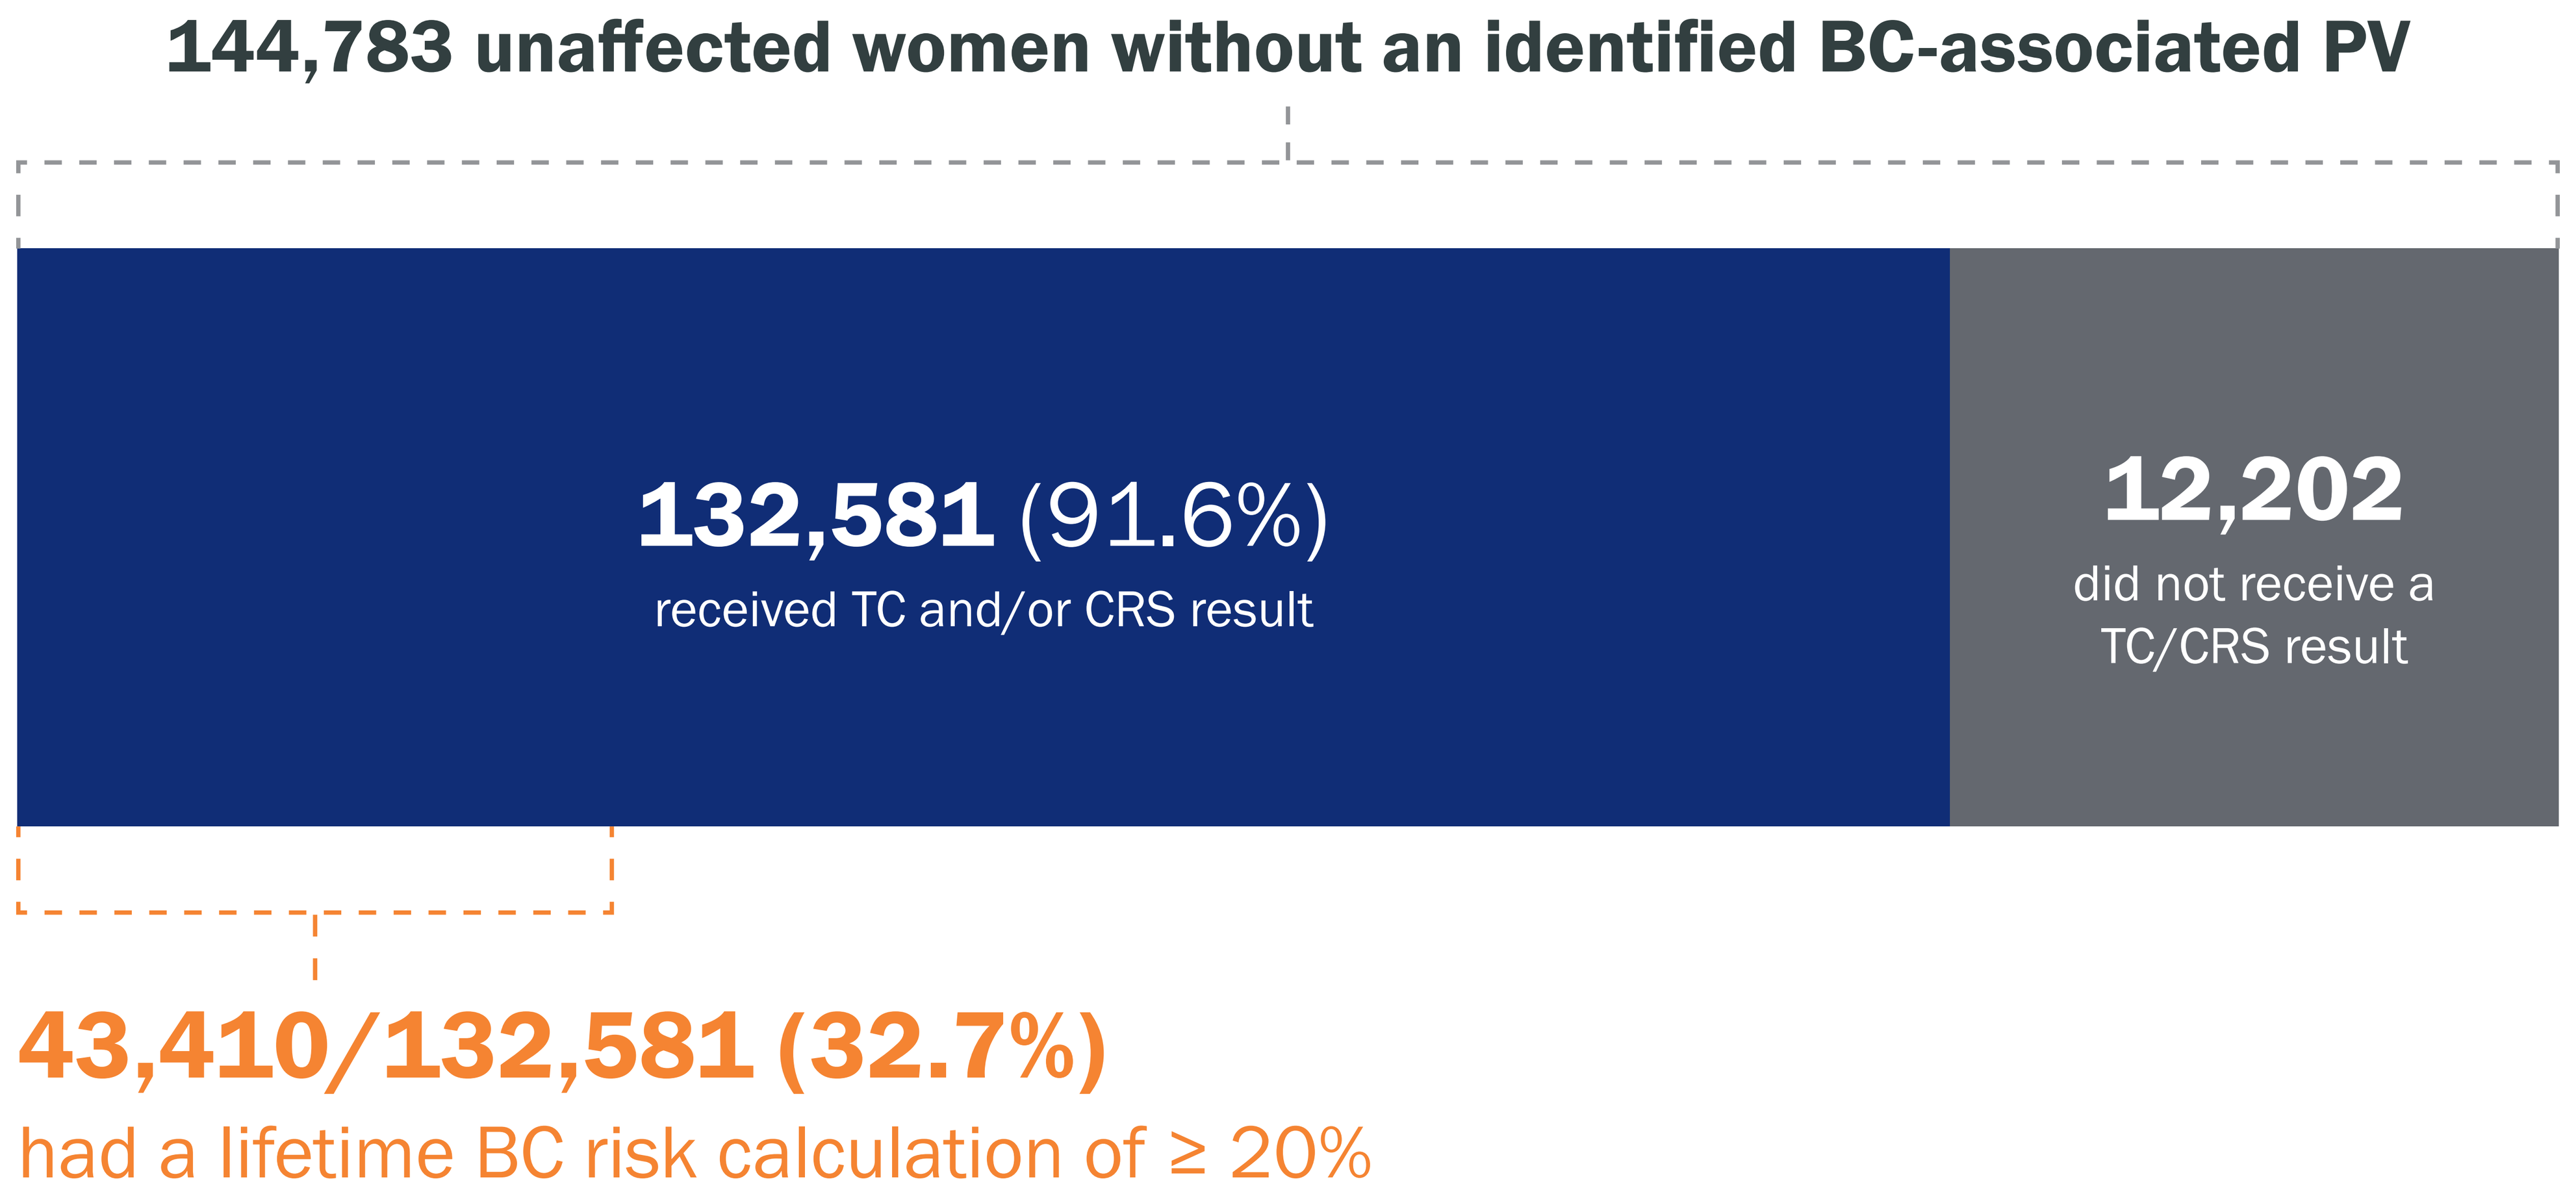
144,783 unaffected women without an identified BC-associated PV
132,581 (91.6%)
received TC and/or CRS result
12,202
did not receive a TC/CRS result
43,410/132,581 (32.7%)
had a lifetime BC risk calculation of ≥ 20%

- 150,604 unaffected women ages 50-60 received genetic testing
- 5,821/150,604 (3.9%) carried a BC-associated PV (**Figure 2**)
- 144,783/150,604 (96.1%) did not carry a BC-associated PV (**Figure 2**)
- 132,581/144,783 (91.6%) received a TC and/or CRS result (**Figure 3**)
- 43,410/132,581 (32.7%) had a lifetime BC risk calculation of ≥ 20% (**Figure 3**)

Conclusions

- In this clinically selected cohort who met guidelines for hereditary cancer testing, a substantial percentage of individuals at ages associated with usual menopause onset and in the years soon after, in which HT decision-making predominantly takes place, were at high risk for BC.
- While HT is an effective strategy to manage menopausal symptoms, it may increase BC risk.
- Assessing breast cancer risk is essential and should be undertaken prior to making HT decisions.
- In individuals identified at elevated risk, caution should be taken when prescribing HT and be accompanied by a detailed discussion of HT risks/benefits with consideration of alternative remedies.

All authors were employees of Myriad Genetics, Inc. at the time of this study and received salaries and stock as compensation.

edith.smith@myriad.com

Poster ID: I-2026-7