Combined prenatal cell-free DNA screening can improve the identification of at-risk pregnancies compared to traditional carrier screening ordered by OB/Gyns

Summer Pierson, MS, CGC; Heather LaBreche, PhD; Seyedmehdi Shojaee, PhD; Lauren Eisemann, MS, CGC; Cathy Wicklund, MS, CGC; Dale Muzzey, PhD; Katie Johansen Taber, PhD

1. Myriad Genetics, Salt Lake City, Utah

Background

- The goal of carrier screening is to identify patients at increased risk of having offspring affected by genetic conditions to enable informed reproductive decision making.
- Guidelines[1] have evolved from recommending screening for only cystic fibrosis (CF) and spinal muscular atrophy (SMA) to discussing expanded carrier screening (ECS) as an acceptable strategy.
- Barriers to offering ECS may include:
 - Inconsistent insurance coverage[2]
 - Limited clinician experience with ECS[3]
 - Ambiguity in guidelines[4]
- These barriers can lead to lower uptake of ECS and fewer informed patients.
- Recent advances in screening have allowed for simultaneous assessment of maternal and fetal recessive disease carrier status, including for CF and SMA, along with fetal aneuploidy and serological compatibility.
- This study aimed to demonstrate that use of this integrated screening approach would identify more at-risk pregnancies than would be identified when ECS is not performed.

Methods

- Retrospective analysis of orders placed for carrier screening from a commercial laboratory (Foresight, Myriad Genetics) between 07/01/2024 and 06/30/2025
 - Orders stratified by clinician type
 - Percentage of OB/Gyns ordering only CF/SMA was calculated
- Retrospective analysis of results from a combined screening assay (FirstGene, Myriad Genetics) that includes 10 recessive conditions, including CF (*CFTR*) and SMA (*SMN1*), between 06/02/2025-03/30/2026 (**Figure 1**)
 - Determined which conditions would have been missed if traditional carrier screening had occurred for only CF and SMA

Figure 1. FirstGene Screen Conditions

1 person

1 screen

FirstGene prenatal cfDNA screening
Prenatal cfDNA aneuploidy screening:
- T21, T18, T13, 22q, XX, XY, X, XXX, XXY, XYY

Fetomaternal blood compatability:
- RHD

Maternal carrier screening:
- Core guidance genes: *CFTR, SMN1*
- Hemoglobinopathies: *HBB, HBA1/2*
- Expanded content: *HEXA, PMM2, ACADM, ASPA, DHCR7, PAH*
- Maternal-only: *FMR1* (via capillary electrophoresis)

Fetal recessive disease status (affected, carrier, or normal):
- Same genes as maternal carrier screening (except *FMR1*)

Outcomes

Table 1. Pregnancies identified at increased risk for recessive conditions in the first 10 months of combined screening assay availability.

Recessive Gene	No. of pregnancies at increased risk
Hb beta chain-related hemoglobinopathy (*HBB*)	3
Cystic Fibrosis (*CFTR*)	3
Smith-Lemli-Opitz syndrome (*DHCR7*)	1
Congenital disorder of glycosylation type 1a (*PMM2*)	1

Increased risk is defined as a pregnancy in which at least one variant was detected within the fetus but the assay was unable to definitively rule out the possibility of two variants for the given recessive condition.

Table 2. Fetal carriers identified in the first 10 months of combined screening assay availability.

Recessive Gene	Fetal carriers
Medium chain acyl-CoA dehydrogenase deficiency (*ACADM*)	19
Canavan disease (*ASPA*)	3
Cystic Fibrosis (*CFTR*)	38
Smith-Lemli-Opitz syndrome (*DHCR7*)	26
Hb beta chain-related hemoglobinopathy (*HBB*)	32
Hexosaminidase A deficiency (including Tay-Sachs disease) (*HEXA*)	4
Phenylalanine hydroxylase deficiency (*PAH*)	29
Congenital disorder of glycosylation type 1a (*PMM2*)	10
Spinal Muscular Atrophy (*SMN1*)	10

- 26% of carrier screening orders placed by OB/Gyn providers were a limited panel of only two conditions, CF and SMA.
- In the first 10 months since the combined screening assay became available, 8 pregnancies were identified at increased risk for recessive conditions (**Table 1**).
- Fetal carriers were identified in 171 samples (**Table 2**). In these patients, if only CF and SMA carrier screening had been performed, 5 pregnancies at increased risk and 123/171 fetal carriers would have been missed.

Implications for Women's Health

- Traditional carrier screening for CF and SMA will miss pregnancies at risk for other recessive conditions.
- For clinicians who only order CF and SMA or another limited panel, an integrated and simplified screening platform that performs multiple analyses simultaneously may increase the detection of at-risk pregnancies and provide more equitable care.

1. Committee Opinion No. 690 Summary: Carrier Screening in the Age of Genomic Medicine. Obstet Gynecol. 2017 Mar;129(3):595-596. doi: 10.1097/AOG.0000000000001947. PMID: 28225420. **2.** Green S, Hartzfeld D, Terry AB, Fissell K, Friedman S, Paolino N, Principe K, Sandbach J, Trzupek K, Winheld S, Malinowski J. An evidence-based practice guideline of the National Society of Genetic Counselors for telehealth genetic counseling. J Genet Couns. 2023 Feb;32(1):4-17. doi: 10.1002/jgc4.1627. Epub 2022 Aug 30. PMID: 36054686. **3.** Kraft SA, Duenas D, Wilfond BS, Goddard KAB. The evolving landscape of expanded carrier screening: challenges and opportunities. Genet Med. 2019 Apr;21(4):790-797. doi: 10.1038/s41436-018-0273-4. Epub 2018 Sep 24. PMID: 30245516; PMCID: PMC6752283. **4.** Pilenzi L, Scorrano V, Di Rado S, Buccolini C, Giansante R, Siciliani L, Stuppia L, Gatta V, Capalbo A. Expanded Carrier Screening: Current Evidence and Future Directions in the Era of Population Genomics. Genes (Basel). 2026 Jan 5;17(1):58. doi: 10.3390/genes17010058. PMID: 41595478; PMCID: PMC12841374.

All authors were employees of Myriad Genetics, Inc. at the time of this study and received salaries and stock as compensation.

Email for questions: summer.pierson@myriad.com

Poster ID: I-2026-5