POSTER #4

Age and Ancestry-Specific Prevalence of Pathogenic Variants in Individuals with Endometrial Cancer Who Do Not Meet Guidelines for Hereditary Cancer Testing: an Opportunity to Reduce Health Disparities

Edith Smith, DNP, CNM, WHNP-BC, AGN-BC; Ryan Bernhisel, MStat; Matthew Kucera, MS; Elisha Hughes, PhD

Myriad Genetics, Inc., Salt Lake City, UT

Myriad genetics®

Disclosures: All authors were employees of Myriad Genetics, Inc. at the time of this study and received salaries and stock as compensation.

Background

- Uterine cancer is the most prevalent gynecologic malignancy in the US with endometrial carcinoma (EC) accounting for the majority of cases.
- Endometrial proliferation due to unopposed estrogen exposure is the etiology responsible for most EC, though 2-5% are due to inherited (germline) pathogenic variants (PVs).
- Guidelines exist for the identification and testing of individuals at risk for carrying a PV. Oftentimes, however, specific criteria must be met to pursue testing, including younger age at diagnosis, family history (FH), multiple primary cancers, and/or certain tumor characteristics.
- Significant health disparities in cancer incidence, treatment, and outcomes exist among individuals with EC. Many individuals who do not meet guideline criteria may unknowingly harbor PVs and consequently receive suboptimal care.
- While EC is predominantly managed by gynecologic oncology specialists, it is often first identified in the women's health setting.
- Midwives are an integral part of the collaborative care team and are involved in the care of patients with EC, predominantly before/at diagnosis and in the post-treatment survivorship phase, all of which provide opportunities to identify those with hereditary risk.

Objective: We aimed to estimate the age and ancestry-specific prevalence of PVs among individuals with EC who may not meet guidelines for genetic testing.

Methods

- Multivariable logistic regression models were constructed to analyze trends in the prevalence of PVs based on ancestry, age, personal cancer history, and FH in a large clinical cohort of patients referred for hereditary cancer genetic testing (MyRisk, Myriad Genetics).
- Ancestry-specific model-based PV prevalence estimates for patients without FH diagnosed with EC at various ages are reported.
- Overall PV prevalence was summarized among individuals undergoing multigene panel testing for 25 to 63 hereditary cancer genes, including 11 established endometrial cancer susceptibility genes.

Results

- The study cohort comprised 26,403 individuals affected with EC. Self-reported ancestries are listed in **Figure 1**.
- The age group with the highest prevalence of PVs for all genes and genes most frequently implicated in EC was ages 50-54 (**Tables 1 and 2**).
- PV prevalence estimates remained consequential for those aged 70+ for all genes (4.6%) and genes most frequently implicated in EC (1.6%) (**Table 1**).
- Overall PV prevalence among individuals with EC but without a FH of cancer was estimated to be 7.3%. Asian individuals had the highest estimated prevalence for all genes (10.5%) and those most frequently implicated in EC (8.2%) (**Table 2**).

Figure 1. Cohort ancestry

Ancestry	N (%)
Asian	677 (2.6%)
Black	1,819 (6.9%)
Hispanic	3,059 (11.6%)
White	15,296 (57.9%)
Multiple	1,935 (7.3%)
Other	670 (2.5%)
Unknown	2,947 (11.2%)

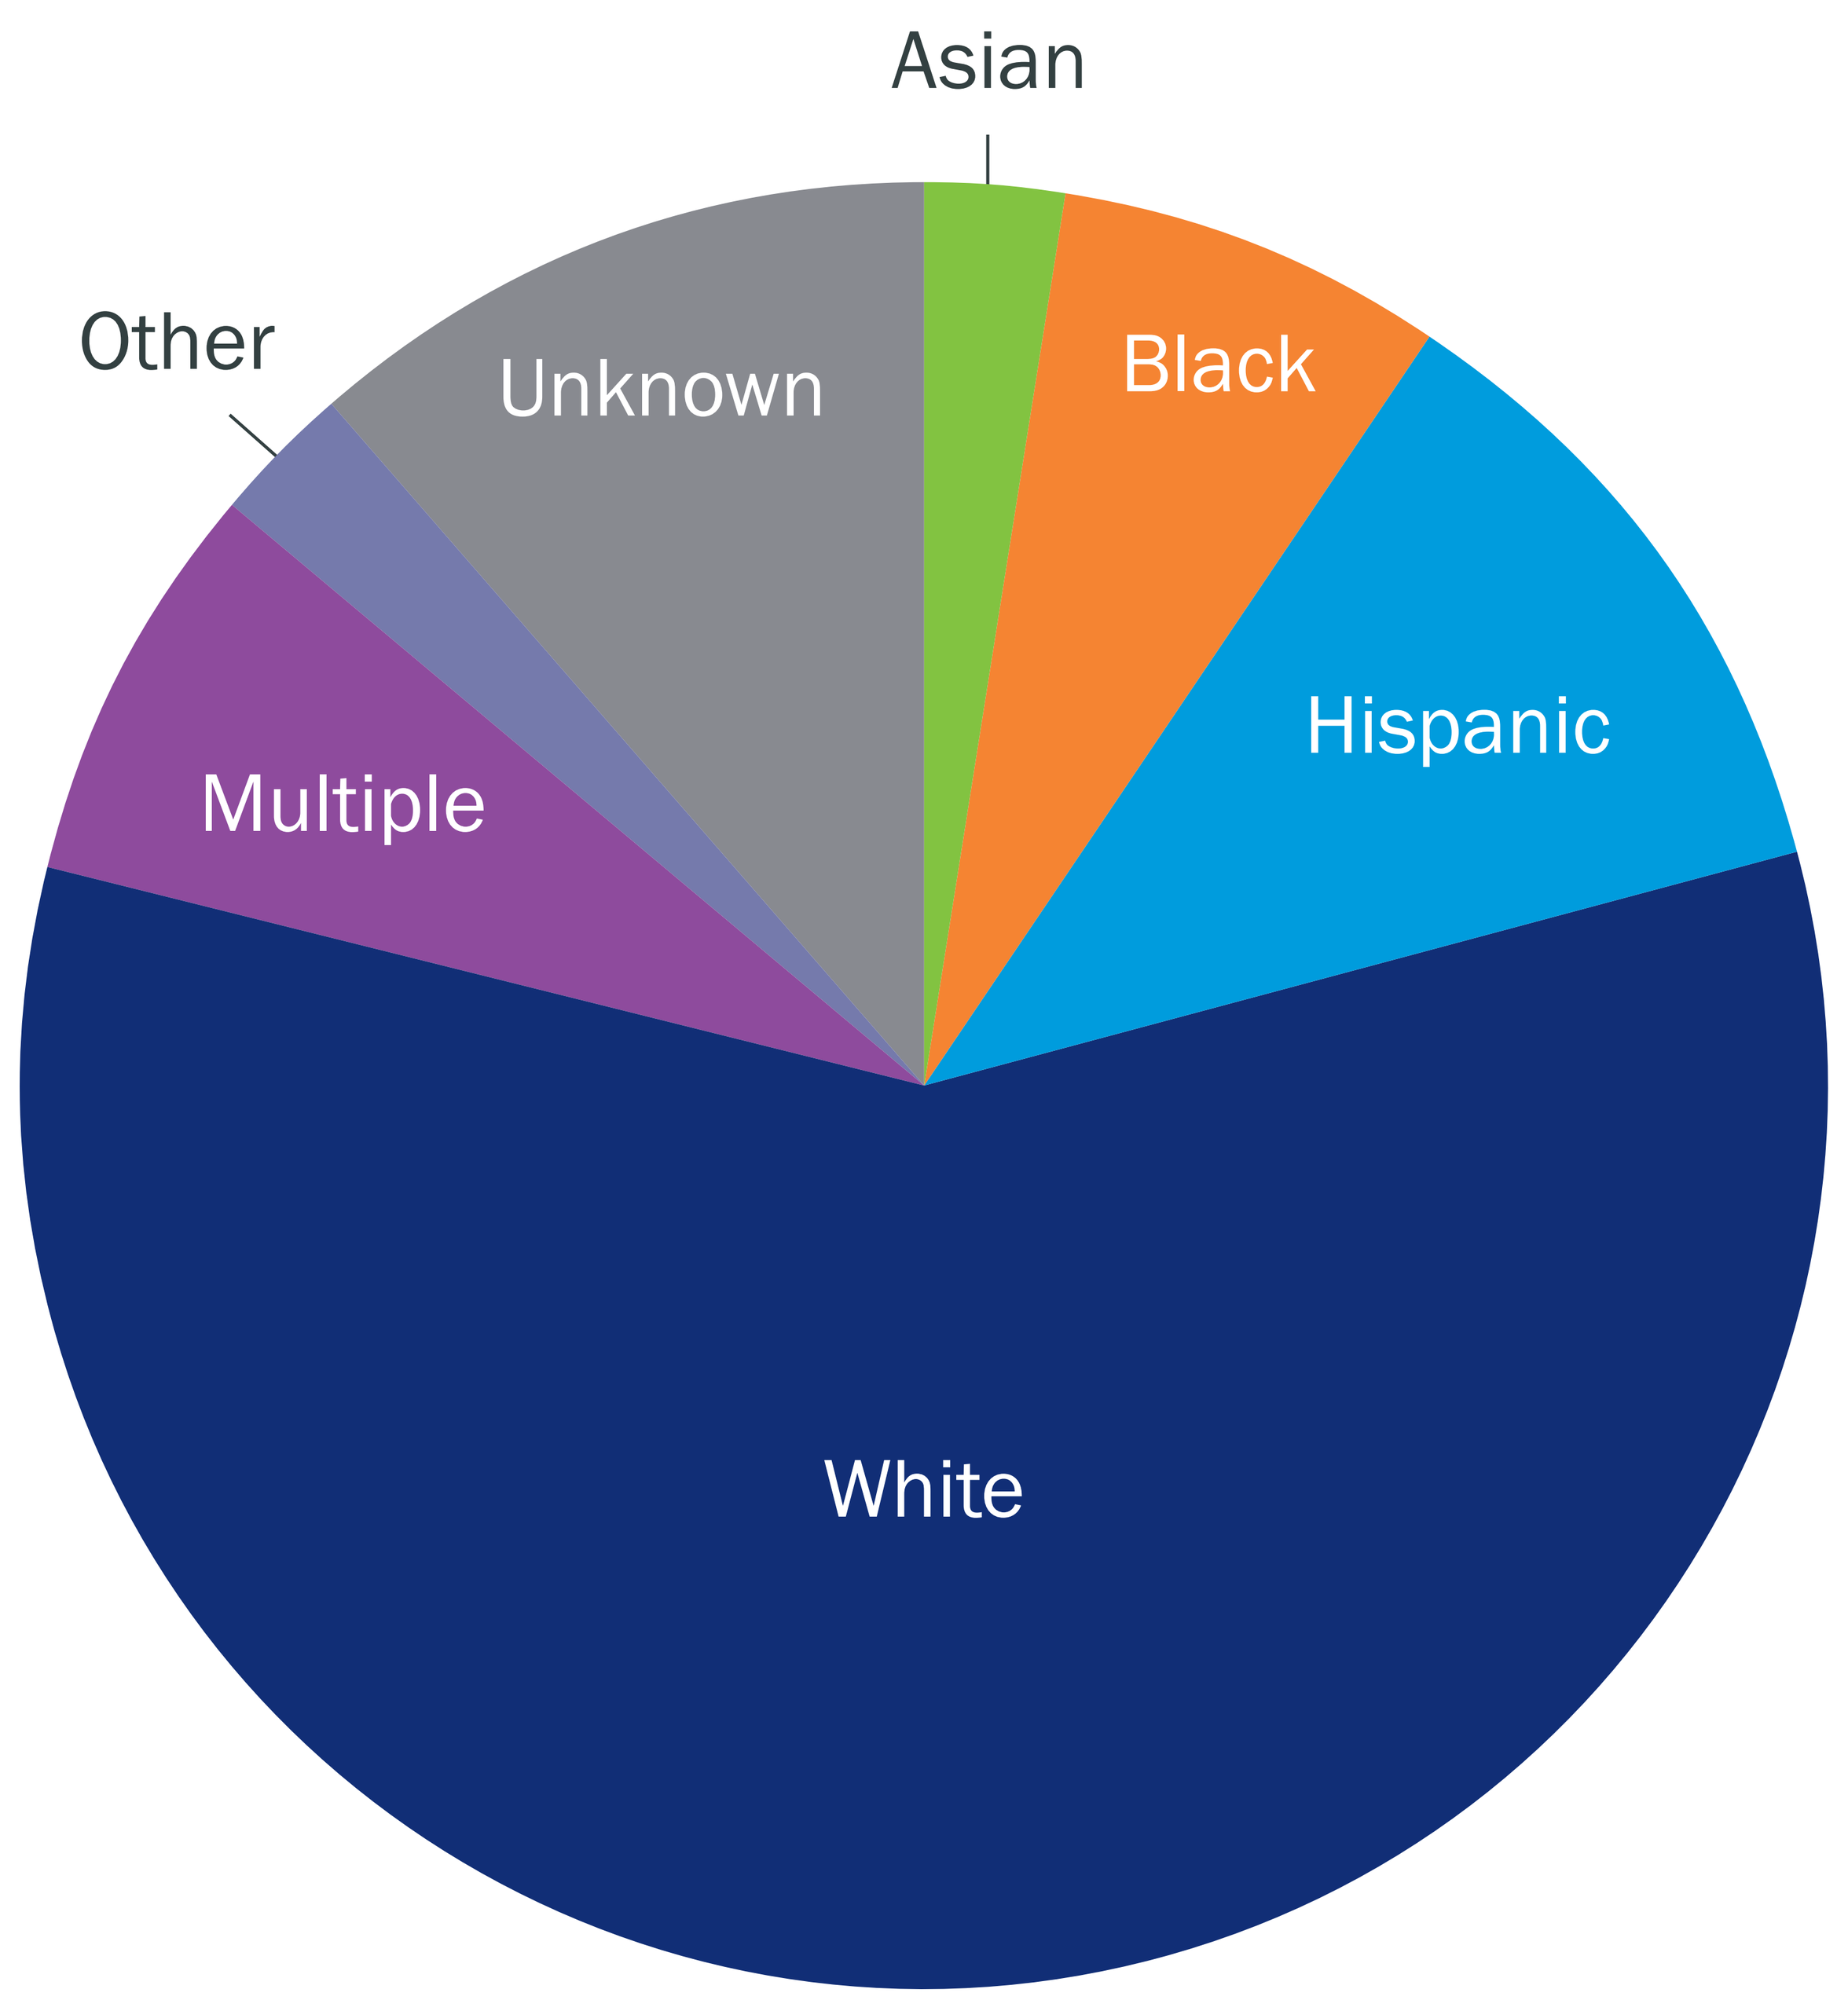
Asian
Black
Hispanic
White
Multiple
Other
Unknown

The study cohort comprised 26,403 individuals affected with EC. Self-reported ancestries are listed. The "Other" category comprised Native American (N=208), Ashkenazi Jewish (N=190), Middle Eastern (N=74), Pacific Islander (N=67), and Other ancestries (N=131).

Table 1. Age of EC diagnosis and estimated PV prevalence (%)

Genes	<20	20-24	25-29	30-34	35-39	40-44	45-49	50-54	55-59	60-64	65-69	70+
All Genes	2.7	3.2	3.7	4.9	6.4	8.8	10.0	11.2	8.7	5.2	4.8	4.6
EC Genes	0.0	0.4	0.9	2.2	2.9	5.8	6.8	7.3	5.2	2.2	1.6	1.6

EC-associated genes: *MLH1, MSH2, MSH6, PMS2, EPCAM, POLE, POLD1, PTEN, NTHL1, STK11, TP53.*

Table 2. PV prevalence by ancestry for all ages and ages 50-54

	All ages		Ages 50-54	
Ancestry	All genes %	EC genes %	All genes %	EC genes %
All ancestries	7.3%	4.1%	11.2%	7.3%
Asian	10.5%	8.2%	14.4%	12.0%
Black	5.6%	3.4%	9.5%	7.0%
Hispanic	7.1%	3.7%	10.8%	6.4%
White	7.4%	4.7%	11.4%	8.4%
Multiple ancestries	7.3%	3.4%	11.2%	5.8%

EC-associated genes: *MLH1, MSH2, MSH6, PMS2, EPCAM, POLE, POLD1, PTEN, NTHL1, STK11, TP53.*

Conclusions

- Among all ancestries, a significant proportion of individuals with EC who do not meet genetic testing guidelines may carry PVs.
- The highest prevalence of PVs was in individuals ages 50-54.
- Removing the barrier of age-based testing for genetic evaluation may positively impact outcomes, especially among disadvantaged groups.
- Appropriate identification of individuals with hereditary risk for EC, by midwives and other primary women's healthcare providers, presents a tremendous opportunity to address disparate care among those most vulnerable.

Email for questions: edith.smith@myriad.com